Alternative Topical Treatment to an Aluminum Chloride Antiperspirant that Provides Prescription Strength Efficacy with Significantly Less Irritation

Mike Thomas BS, MBA, Lola Kelly Smalls PhD, MBA, Tyra Leazer PhD, Elizabeth Finn PhD, Laurie Elstun M.D., Paula Hartwig BS, David Swaile PhD, Amy Capretta BS

INTRODUCTION

As reported at the 2006 AAD Annual Meeting, patients with severe axillary sweating (including hyperhidrotics) struggle daily with the condition, limiting their relationships, careers, and aspirations. Prescription Strength (Rx) and OTC aluminum chloride antiperspirants are frequently a recommended treatment for these patients. While this course of treatment is often very efficacious, many patients develop irritation that is sufficiently severe and can lead to discontinued use.

OBJECTIVE

To demonstrate that nighttime underarm application of an OTC anhydrous "soft solid" antiperspirant (AP) containing a non-Aluminum Chloride active ingredient (Aluminum Zirconium Trichlorohydrex), can deliver statistically similar efficacy to a prescription AP containing aluminum chloride while providing a substantial decrease in patient axillary irritation.

METHODS

Clinical Test Protocol

Two clinical studies were designed to quantify the underarm wetness performance of an OTC anhydrous AP relative to a marketed prescription AP (6% Concentration). The panelists had product applied to their underarms (product application was randomized and blinded) every evening for 7 days (In the second study, the duration was extended to 10 days). Baseline sweat collection occurred on Day 1 before product was applied and another sweat collection occurred on Day 7. (In the second study, an additional measurement was taken at Day 10). The panelists were evaluated for irritation during each visit. If a panelist had an irritation score of 2 or above, product application was discontinued and the highest irritation score for that panelist was carried forward for the analysis. Even if application was discontinued, all subjects were eligible to participate in sweat collection regardless of treatment compliance and were considered in the analyses to provide an estimate representative of actual practice. Note that only the Prescription Aluminum Chloride AP usage led to an irritation score of 2 or above and required discontinuation.

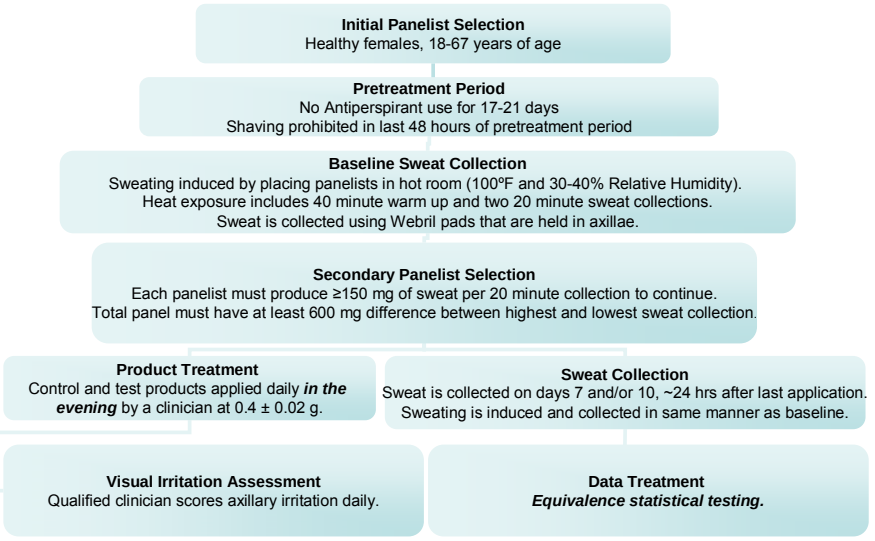
Initial Panelist Selection
Healthy females, 18-67 years of age
Pretreatment Period
No Antiperspirant use for 17-21 days
Shaving prohibited in last 48 hours of pretreatment period
Baseline Sweat Collection
Sweating induced by placing panelists in hot room (100ºF and 30-40% Relative Humidity).
Heat exposure includes 40 minute warm up and two 20 minute sweat collections.
Sweat is collected using Webril pads that are held in axillae.
Secondary Panelist Selection
Each panelist must produce ≥150 mg of sweat per 20 minute collection to continue.
Total panel must have at least 600 mg difference between highest and lowest sweat collection.
Product Treatment
Control and test products applied daily *in the evening* by a clinician at 0.4 ± 0.02 g.
Sweat Collection
Sweat is collected on days 7 and/or 10, ~24 hrs after last application.
Sweating is induced and collected in same manner as baseline.
Visual Irritation Assessment
Qualified clinician scores axillary irritation daily.
Data Treatment
Equivalence statistical testing.

Data Analysis

Mixed model techniques were used to analyze the gravimetric sweat data and a 95% confidence interval was constructed to assess non-inferiority. A boundary condition of 20% was used for the non-inferiority criteria because in a controlled, single product blind test study placed among females, consumers did not notice differences in level of sweat between products with clinically demonstrated differences of greater than 20%.

RESULTS

Clinical Efficacy Results

• In both clinical trials, the OTC anhydrous "soft solid" AP (Treatment A) provided statistically similar efficacy to the Prescription Aluminum Chloride AP (Treatment C).

Clinical Study #1, n=19

Evaluation	Trt	Adjusted Mean Sweat Collected (mg)	Percent Difference {(SS-Rx)/SS}*100% [Confidence Bound]*
Post Trt 6	A	175	2.6%** [18.5%]
	C	170	

*For both studies, a confidence bound less than 20% indicates that Treatment A is non-inferior to Treatment C.

Clinical Study #2, n=29

Evaluation	Trt	Adjusted Mean Sweat Collected (mg)	Percent Difference {(SS-Rx)/ SS}*100% [Confidence Bound]*
Post Trt 6	A	141	-12.7%** [-1.6%]
	C	159	
Post Trt 9	A	173	-5.7%** [5.1%]
	C	182	

**Calculations based on log10 transformed data

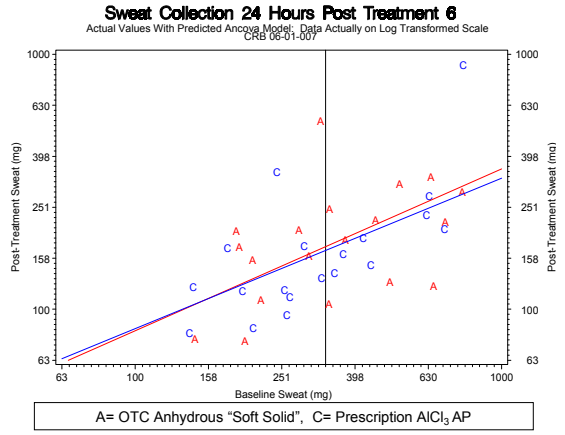
Sweat Collection 24 Hours Post Treatment 6
Actual Values With Predicted Ancova Model: Data Actually on Log Transformed Scale
CRB 06-01-007
Post-Treatment Sweat (mg)
Baseline Sweat (mg)
A= OTC Anhydrous "Soft Solid", C= Prescription $AlCl_3$ AP

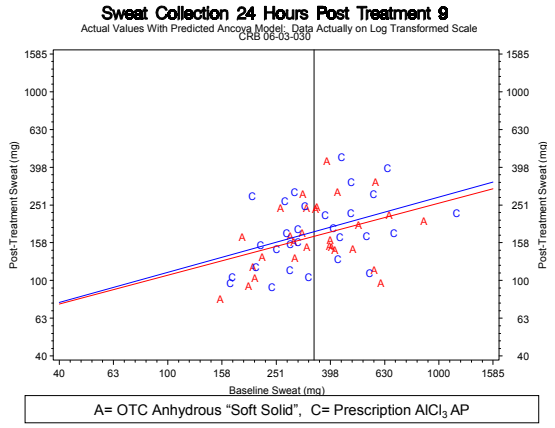
Sweat Collection 24 Hours Post Treatment 9
Actual Values With Predicted Ancova Model: Data Actually on Log Transformed Scale
CRB 06-03-030
Post-Treatment Sweat (mg)
Baseline Sweat (mg)
A= OTC Anhydrous "Soft Solid", C= Prescription $AlCl_3$ AP

Skin Irritation Results

• In both clinical trials, the OTC anhydrous "soft solid" AP provided a statistically significant advantage to the Prescription Aluminum Chloride AP.

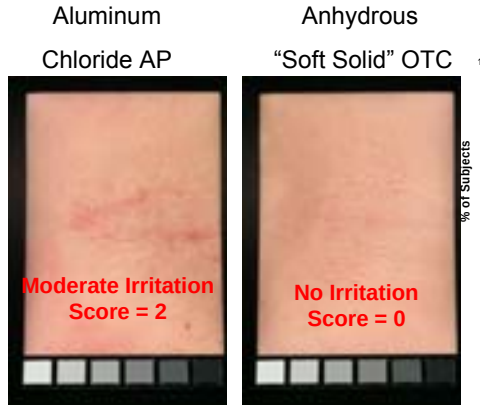
Aluminum Chloride AP
Anhydrous "Soft Solid" OTC
Moderate Irritation Score = 2
No Irritation Score = 0

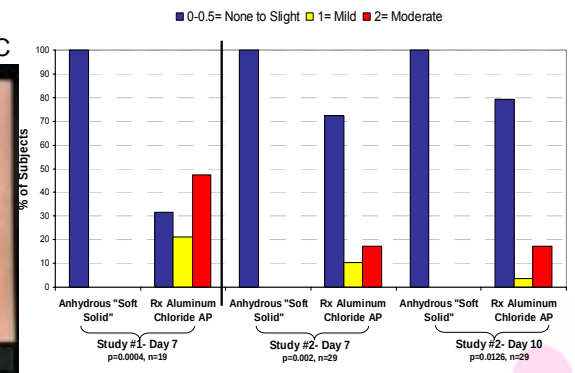
0-0.5= None to Slight
1= Mild
2= Moderate
% of Subjects
Anhydrous "Soft Solid"
Rx Aluminum Chloride AP
Study #1- Day 7
p=0.0004, n=19
Study #2- Day 7
p=0.002, n=29
Study #2- Day 10
p=0.0126, n=29

CONCLUSIONS

- **Overall, nighttime application of an OTC anhydrous AP containing an Aluminum Zirconium Trichlorohydrex Gly active ingredient does deliver statistically similar efficacy to a Prescription Aluminum Chloride AP.**
- **Additionally, the "soft solid" AP demonstrates the ability to deliver this efficacy without the key compliance negative (irritation) of current Prescription Aluminum Chloride APs.**

References

1. Thomas M, Pieretti L, Swaile D, Capretta A, Hill T, Panel Discussions Among Hyperhidrosis Patients to Assess Lifestyle Limitations, Treatment Regimes, and Compensating Behaviors. American Academy of Dermatologists Annual Meeting. 2006.

2. Swaile DF, Chabi GH, Putman CM. Clinical Comparison of Antiperspirant Efficacy as a Function of Morning and/or Nighttime Application. American Academy of Dermatologists Annual Meeting. 2005.

This work was funded by P&G Beauty